5th International Congress on Drug Therapy in HIV Infection, Glasgow, UK (October 22-26, 2000)

Durable Suppression of HIV+ RNA After Two Years of Kaletra (ABT-378/ritonavir) Therapy in Single Protease Inhibitor Experienced Patients

J. Feinberg[1], S. Brun[14], T. Marsh[14], M. King[14], C. Benson[2], S. Deeks[3], H. Kessler[4], R. Murphy[5], R. Gulick[6], D. Wheeler[7], C. Hicks[8], J. Eron[9], P. Sax[10], R. Stryker[11], S. Riddler[12], M. Thompson[13], K. Real[14], A. Hsu[14], R. Bertz[14], D. Kempf[14], A. Japour[14], and E. Sun[14] for the M97-765 Study Group; [1]U. Cincinnati, [2]U. Colorado, [3]UC-SF, [4]Rush, [5]Northwestern, [6]Cornell, [7]Infectious Diseases Physicians, [8]Duke, [9]U. N. Carolina, [10]Harvard, [11]Pacific Oaks Research, [12]U. Pittsburgh, [13]AIDS Research Consortium of Atlanta, and [14]Abbott Laboratories

BACKGROUND

ABT-378 (lopinavir) is a novel HIV protease inhibitor (PI) that is co-formulated with ritonavir, an inhibitor of cytochrome P450 3A. It is exquisitely sensitive to pharmacokinetic enhancement by ritonavir, resulting in substantially increased ABT-378 drug exposure, even at low ritonavir doses. At the dosage selected for phase III clinical trials, 400 mg ABT-378/100 mg ritonavir BID (as 3 co-formulated capsules BID), ritonavir concentrations are below those required for antiviral activity.[1] The mean ABT-378 C_{trough}/EC_{50} ratio (Inhibitory Quotient or IQ) for wild-type HIV is ≥75 when dosed at 400/100 mg BID, potentially providing a pharmacologic barrier to the emergence of viral resistance and activity against resistant virus.[1] In contrast, the IQs for currently available PIs whether dosed as single PIs or in conjunction with ritonavir, range from 1-26 (Figure 1), based on the EC_{50} values determined in the presence of 50% human serum.[2] C_{trough} values in HIV+ patients for currently available PIs are derived from published sources including package inserts.[3-14]

The efficacy and safety of Kaletra (lopinavir/ritonavir, formerly known as ABT-378/r) are currently being studied in HIV-infected patients, both antiretroviral-naïve and PI-experienced. The M97-765 study is an ongoing phase II trial of ABT-378/r in combination with nevirapine and 2 NRTIs in single PI-experienced, NNRTI-naïve patients. Results through Week 96 are presented here.

Figure 1. C_{trough}/EC_{50} Ratio (Inhibitory Quotient) for PIs Given Alone or with RTV*

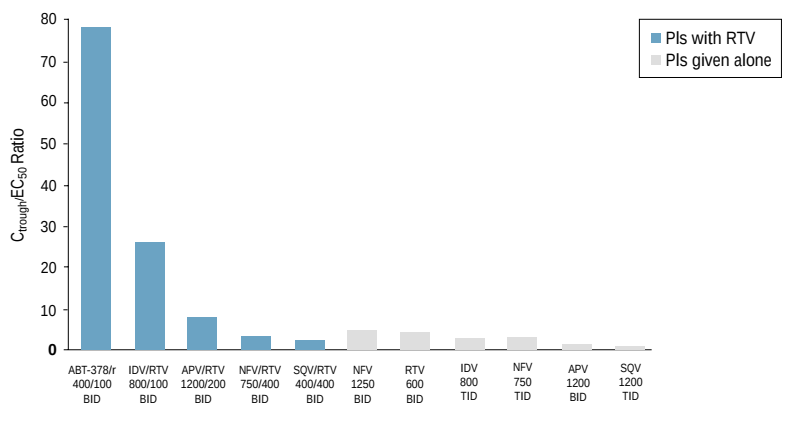
PIs with RTV
PIs given alone
C_{trough}/EC_{50} Ratio
ABT-378/r 400/100 BID
IDV/RTV 800/100 BID
APV/RTV 1200/200 BID
NFV/RTV 750/400 BID
SQV/RTV 400/400 BID
NFV 1250 BID
RTV 600 BID
IDV 800 TID
NFV 750 TID
APV 1200 BID
SQV 1200 TID

* Contribution of RTV to antiviral activity not included; regimens shown are those where data in HIV-infected patients are available; EC_{50} values adjusted for protein binding.

METHODS

Entry Criteria

- Plasma HIV RNA 10^3–10^5 copies/mL.
- On treatment with 1 PI and 2 NRTIs for ≥3 months at study entry.
- Single PI-experienced (no prior treatment with any PI, other than the current one, for ≥6 weeks, and no prior dual PI therapy).
- Naïve to at least one NRTI, with no prior NNRTI experience.

Study Design and Analysis

- Seventy patients were randomized to receive ABT-378/r (400/100 mg BID or 400/200 mg BID) in place of their current PI, in combination with their existing NRTIs.
- On study day 15, nevirapine (200 mg QD for 14 days then 200 mg BID) was added and NRTIs were changed to include at least one which was new to the patient (Figure 2).
- Plasma HIV RNA was quantified using Roche Amplicor HIV-1 Monitor™ (lower limit of quantitation [LLQ] 400 copies/mL) and the Roche Amplicor HIV-1 Monitor Ultrasensitive HIV RNA quantitative PCR, version 1.0 (LLQ 50 copies/mL).

Figure 2. M97-765 Study Schema

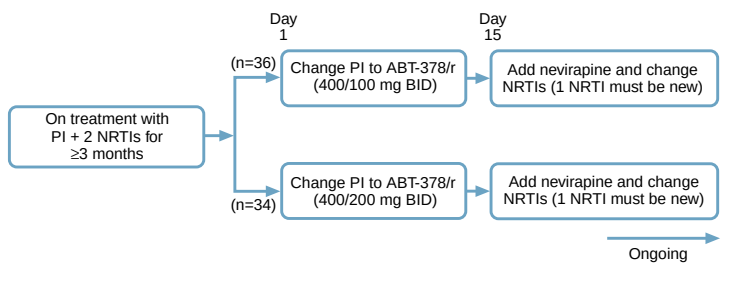
Day 1
Day 15
On treatment with PI + 2 NRTIs for ≥3 months
(n=36)
Change PI to ABT-378/r (400/100 mg BID)
Add nevirapine and change NRTIs (1 NRTI must be new)
(n=34)
Change PI to ABT-378/r (400/200 mg BID)
Add nevirapine and change NRTIs (1 NRTI must be new)
Ongoing

RESULTS

Baseline Characteristics

- Sixty-three male and 7 female patients: 66% Caucasian, 24% Black, 7% Hispanic, 3% Asian/Pacific Islander.
- Mean age: 40 years (range 22–66).
- Median plasma HIV RNA: 4.0 $\log_{10}$ copies/mL (range 2.9–5.8).
- Median CD4 count: 349 cells/mm³ (range 72–807).
- Previous PI-experience included indinavir (44%), nelfinavir (36%), saquinavir (13%), ritonavir (6%), and amprenavir (1%).
- 3TC was the most common NRTI being used at baseline (87%), followed by d4T (56%) and zidovudine (43%), with a small minority receiving ddI (10%).

Baseline Viral Susceptibility

- Protease inhibitor phenotypic susceptibility data were available for 57/70 baseline viral isolates (VIRCO Antivirogram® method).
- Baseline viruses from 63% of patients (36/57) displayed a ≥4-fold loss in susceptibility to the previous PI, and 32% of patients (18/57) had viruses with ≥4-fold loss in susceptibility to three or more PIs (Table 1).
- Of 56 patients for whom baseline phenotype for all of the drugs in the previous regimen was available, the incidence of ≥4-fold change in EC_{50} to one, two or three previous drugs was 36%, 45%, and 16%, respectively.
- Nineteen percent of patients (11/57) had ≥4-fold reduction in susceptibility to ABT-378 at baseline.

Table 1. Baseline Susceptibility to Previous Protease Inhibitor

		Fold Change in EC_{50} Compared with Wild-Type	
Previous PI	n	Mean	Range
Indinavir	24	7.4	0.4–26
Nelfinavir	21	19.1	0.8–>54
Saquinavir	9	9.5	0.7–32
Ritonavir	3	23.0	7.3–41

Viral Load Response at Week 2

- Viral suppression was rapid with a marked reduction in HIV RNA during the first two weeks of therapy. At Week 2, prior to the addition of nevirapine, 80% (56/70) of patients had a ≥1 $\log_{10}$ copies/mL decrease in HIV RNA or achieved a viral load <400 copies/mL (Figure 3).
- Viral load decline through Week 2 was independent of the EC_{50} of ABT-378 against baseline viral isolates (range 0.7–26-fold)(Figure 4). This initial decline was also independent of prior PI experience.

Figure 3. HIV RNA Mean Change from Baseline

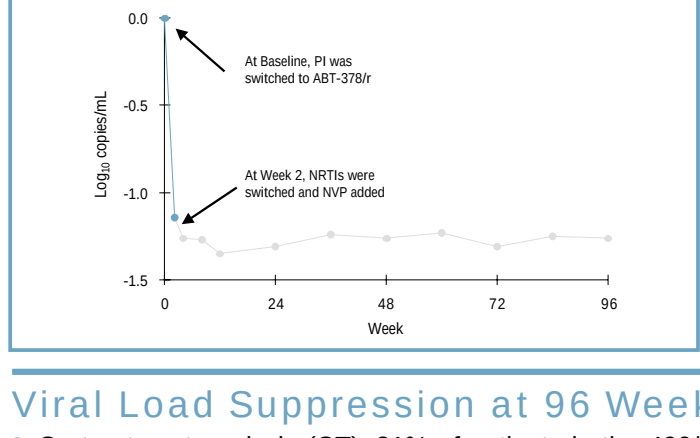
At Baseline, PI was switched to ABT-378/r
At Week 2, NRTIs were switched and NVP added
$\log_{10}$ copies/mL
Week

Figure 4. HIV RNA Decline as a Function of Fold Change in ABT-378 EC_{50}

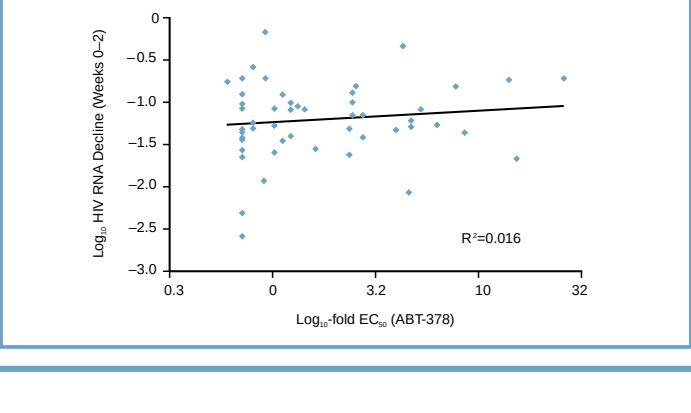
$\log_{10}$ HIV RNA Decline (Weeks 0–2)
R^2=0.016
$\log_{10}$-fold EC_{50} (ABT-378)

Viral Load Suppression at 96 Weeks

- On-treatment analysis (OT): 81% of patients in the 400/100 mg dose group and 81% of patients in the 400/200 mg dose group had viral load (VL) <400 copies/mL at Week 96 (Figure 5).
- Intent-to-treat analysis (ITT M=F; missing values considered as treatment failure): 61% of patients in the 400/100 mg dose group and 65% of patients in the 400/200 mg dose group had a VL <400 copies/mL at Week 96 (Figure 6).

Figure 5. M97-765 HIV RNA<400 Copies/mL (OT)

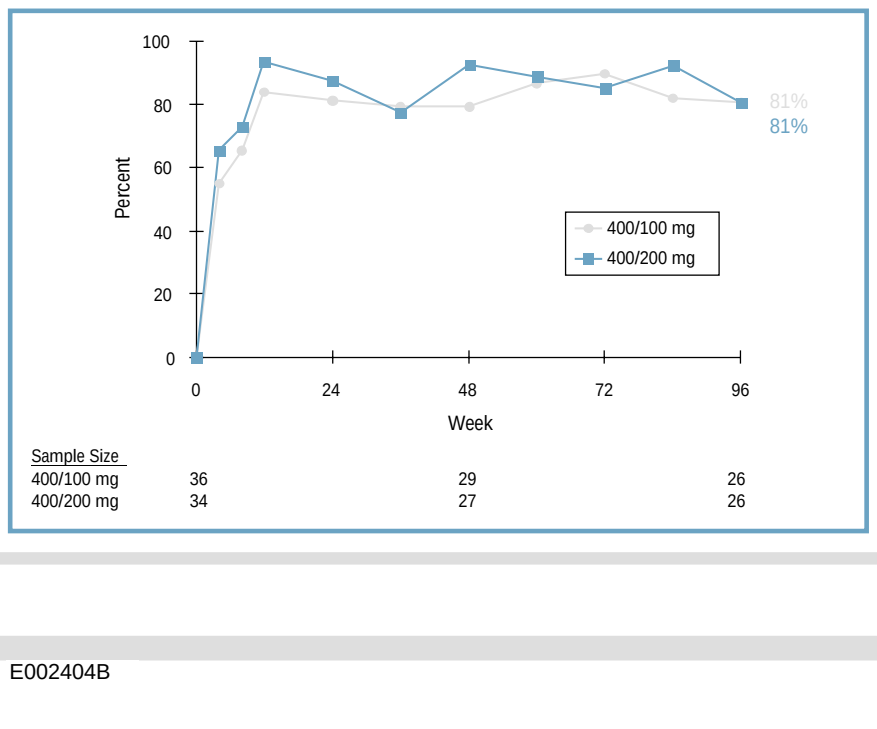
81%
81%
400/100 mg
400/200 mg
Percent
Week
Sample Size
400/100 mg 36 29 26
400/200 mg 34 27 26

Figure 6. M97-765 HIV RNA <400 Copies/mL (ITT M=F)

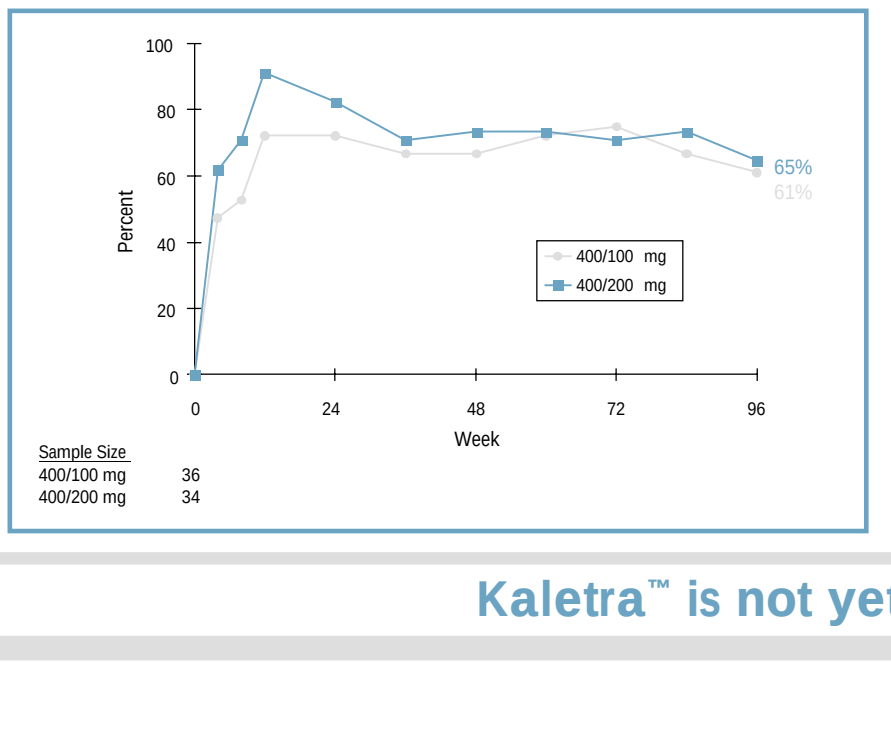
65%
61%
400/100 mg
400/200 mg
Percent
Week
Sample Size
400/100 mg 36
400/200 mg 34

Table 2. Percentage of Patients with Plasma HIV RNA <50 Copies/mL at 96 Weeks

Analysis	400/100 mg (n=36) %	400/200 mg (n=34) %
On-treatment	62%	64%
Intent-to-treat (M=F)	47%	50%

Viral Load Suppression in Patients Discontinuing Nevirapine (NVP)

- 3 patients discontinued NVP within 2-4 Weeks after initiation. All 3 patients remained on study through 96 Weeks and 3/3 had a Week 96 VL <400 copies/mL (3/3 <50 copies/mL).
- 4 additional patients discontinued NVP between Week 12 and Week 72. One patient discontinued the study at Week 36 due to an AE, and the other 3 patients remained on study through 96 Weeks. Two of the 3 patients had a Week 96 VL <400 copies/mL (1/2 <50 copies/mL).

CD4 Cell Response

- The mean CD4 cell count at Week 96 was 575 cells/mm³. (The mean increase from baseline was 188 cells/mm³) (Figure 7).
- Differences between dose groups at Week 96 were not statistically significant.

Figure 7. M97-765 CD4 Cell Count Mean Change from Baseline

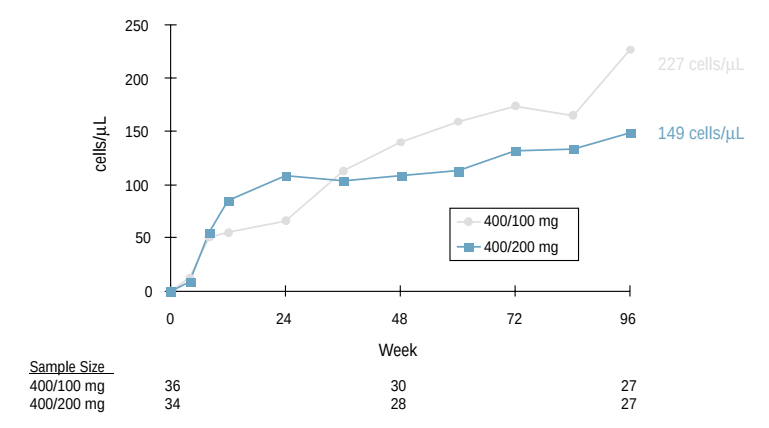
227 cells/µL
149 cells/µL
cells/µL
400/100 mg
400/200 mg
Week
Sample Size
400/100 mg 36 30 27
400/200 mg 34 28 27

Tolerability

- The most common drug-related adverse events through 96 Weeks were diarrhea and asthenia (Table 3).
- Fifteen patients discontinued the study through 96 Weeks; of these, only four discontinuations were due to adverse events related to study drug (Table 4).
- No cases of symptomatic hepatitis attributed to ABT-378/r therapy were reported.

Table 3. Most Common Adverse Events and Laboratory Abnormalities

	400/100 mg (n=36)	400/200 mg (n=34)
Adverse Events*		
Diarrhea⁺	25%	26%
Asthenia	6%	9%
GE Reflux	3%	6%
Nausea	3%	6%
Laboratory Abnormalities		
Glucose (>250 mg/dL)	3%	6%
Cholesterol (>300 mg/dL)	22%	36%
Triglycerides (>750 mg/dL)	22%	30%
GGT (>5X ULN)	22%	36%
AST/ALT (>5X ULN)	11%	24%
Amylase (>2X ULN)	6%	3%

* Adverse events of at least moderate severity and probable, possible, or unknown relationship to ABT-378/r are included
⁺ >3 loose stools/day

Table 4. Patient Disposition Through Week 96

	400/100 mg	400/200 mg
Patients Enrolled	**36**	**34**
Patients Discontinuing before Week 96	**8**	**7**
Discontinuations Related to ABT-378/r (Diarrhea (2), Flatulence, Rash)	**3**	**1**
Other Reasons for Discontinuation		
AE (myocardial infarction on study day 1)	1	0
Death (metastatic lung carcinoma, Rhabdomyolysis with acute renal failure)	0	2
Noncompliance	1	1
Personal Reasons/Other	3	1
Lost to Follow-up	0	2

CONCLUSIONS

- ABT-378/r exhibits a potent and durable antiviral effect through 96 Weeks in PI-experienced patients, with VL <400 copies/mL in 81% of patients on treatment (ITT M=F: 63%) and <50 copies/mL in 63% of patients on treatment (ITT M=F: 49%).
- ABT-378/r was well tolerated with only 4/70 patients discontinuing due to adverse events related to study drug through 96 Weeks.
- Viral suppression was achieved and maintained in the majority of patients, despite high-level baseline resistance to prior PIs and NRTIs.
- Virologic response was demonstrated during the initial two weeks of the study when the only alteration to the baseline regimen was substitution of the previous PI with ABT-378/r. Moreover, of patients who discontinued nevirapine during the course of this study, 5/7 maintained a viral load <400 copies/mL and 4/7 maintained a viral load <50 copies/mL through 96 Weeks of ABT-378/r therapy.
- While significant viral suppression was observed in this study of patients with prior PI and NRTI experience, antiviral activity in antiretroviral-naïve patients receiving ABT-378/r with d4T and 3TC was higher.[15] This suggests that early use of ABT-378/r may be associated with optimal outcome.

ACKNOWLEDGMENTS

AIDS Research Consortium of Atlanta	*Bohn H, Sullivan M, Enstrom T*
Brigham and Women's Hospital	*Koziol C*
Cornell Clinical Trials Unit	*Sarraco T, Stroberg T*
Duke University Medical Center	*Giner J, Harmon L*
Infectious Disease Physicians	*King T*
Northwestern University	*Bruce J, Wong A, Donath P*
Pacific Oaks Research	*Perry B, Walker S*
Rush Presbyterian St Luke's Medical Center	*Narkiewicz E*
San Francisco General Hospital	*Raggett D*
University of Cincinnati	*Black J, Daniel P, Powell T*
University of Colorado Health Sciences Center	*Canmann S, Putnam B, Ray MG*
University of North Carolina at Chapel Hill	*Marcus C, Ngo L*
University of Pittsburgh	*Rosener R*
VIRCO	*Hertogs K, Schel P, Verbiest W*
PPD Development	*McCarley S, Donnelly A, Jackson S, Nicks B*
Abbott Laboratories	*Kempf D, Flanders R, Lindberg M, Yang G, Rode R*

Nevirapine was provided by Boehringer Ingelheim Pharmaceuticals Inc.
Phenotype testing of baseline viral isolates was performed by VIRCO.

REFERENCES

1. Bertz R, Lam W, Brun S, et al. Multiple-dose pharmacokinetics (PK) of ABT-378/ritonavir (ABT-378/r) in HIV+ subjects. 39th Interscience Conference on Antimicrobial Agents and Chemotherapy, San Francisco, USA, 1999 (abstract 0327).
2. Kempf, DJ, Molla A, Hsu A. Protease inhibitors as anti-HIV agents. Antiretroviral Therapy, American Society for Microbiology Press, E DeClerq, editor. In press.
3. Ritonavir Package Insert.
4. Indinavir Package Insert.
5. Amprenavir Package insert.
6. Nelfinavir Package Insert.
7. Saquinavir (Fortavase) Package Insert.
8. Cohen C, et al. Potent and convenient Fortovase™ (SQV) SGC BID regimens in combination with 2 nucleosides or nelfinavir (NFV) plus 1 nucleoside in HIV-1 infected patients. 12th World AIDS Conference, Geneva, Switzerland, 1998, (abstract 12314).
9. Saag M, et al. Saquinavir systemic exposure and safety on once daily administration of Fortovase™ (Saquinavir) soft gel capsules (FTV) in combination with low-dose ritonavir. 39th Interscience Conference on Antimicrobial Agents and Chemotherapy, San Francisco, USA, 1999, (abstract 330).
10. Shulman N, et al. Ritonavir intensification in indinavir recipients with detectable HIV RNA levels. 7th Conference on Retroviruses and Opportunistic Infections, San Francisco, USA (abstract 534).
11. Piscitelli S, et al. The addition of a second protease inhibitor eliminates Amprenavir-Efavirenz drug interactions and increases plasma Amprenavir concentrations. 7th Conference on Retroviruses and Opportunistic Infections, San Francisco, USA (abstract 78).
12. Mallolas J, et al. A dose-finding study of once-daily Indinavir/Ritonavir plus Combivir® in HIV infected patients. First International Workshop on Clinical Pharmacology of HIV Therapy, March 2000 (abstract 2.14).
13. Flexner C, et al. Steady-state pharmacokinetic interactions between ritonavir (RTV), nelfinavir (NFV), and the nelfinavir active metabolite M8 (AG1402), 12th World Aids Conference, Geneva (abstract 42265).
14. Cameron DW, et al. Ritonavir and saquinavir combination therapy for the treatment of HIV infection. AIDS, 1999, 12(2) 213-224.
15. Benson C, King M, Brun S, et al. ABT-378/ritonavir (ABT-378/r) in antiretroviral-naïve HIV+ patients: 96 weeks. 40th Interscience Conference on Antimicrobial Agents and Chemotherapy, Toronto, Canada, Sept. 2000 (Poster 546).

Kaletra™ is not yet licensed in the U.K.

E002404B